Point-of-care CART manufacture and delivery: Expanding access to CART therapy via local institutions, Hadassah Medical Center experience.

M. Assayag[1], N. Asherie*[1], S. Kfir-Erenfeld*[1], B. Avni[1], T. Dubnikov[1], N. Zalcman[1], E. Lebel[2], E. Zimran[2], A. Shaulov[2], M. Pick[2], Y. Cohen[3], I. Avivi[3], C. Cohen[4], M.E. Gatt[2], S. Grisariu[1], P. Stepensky[1]

[1]Department of Bone Marrow Transplantation and Cancer Immunotherapy, Hadassah Medical Center, Faculty of Medicine, Hebrew University of Jerusalem, Jerusalem, Israel, [2]Department of Hematology, Hadassah Medical Center, Faculty of Medicine, Hebrew University of Jerusalem, Jerusalem, Israel, [3]Department of Hematology, Tel Aviv Medical Center, Sackler faculty of medicine, Tel Aviv University, Tel Aviv, Israel, [4]. Laboratory of Tumor Immunology and Immunotherapy, The Mina and Everard Goodman Faculty of Life Sciences, Bar-Ilan University, Ramat Gan, Israel.

INTRODUCTION

Anti-BCMA chimeric antigen receptor (CAR) T-cells for the treatment of multiple myeloma (MM) have proven safe and efficacious. Recently, two commercial products (i.e., ABECMA™ and CARVYKTI™) have been approved by the FDA and EMA agencies, auguring a rapid expansion of these "living-drugs" to the market. However, this is not the case yet, since access to this modality of treatment remains very limited. The lack of production slots, manufacturing costs, and logistical issues are major drawbacks of this therapy and present significant obstacles to meeting the growing demand.

Point-of-care CART manufacture and delivery is an especially attractive approach for boosting the supply of CART products. In Israel, about 500 cases of myeloma are newly diagnosed every year, and about 100 cases will become candidates for CART therapy, as third-line therapy. At Hadassah Medical Center (HMC) we have developed a new anti-BCMA CAR (HBI0101) for the treatment of MM and light chain amyloidosis (AL). CART products are manufactured and delivered to patients in-house, considerably shortening the "vein-to-vein" waiting time, which is critical especially for rapidly deteriorating patients.

RESULTS

The results of the phase Ia/b study (NCT04720313), exploring the safety and efficacy effects in 47 patients (42 MM- and 5 AL- patients) at escalating doses ranging from 150- to 800x10^6 CAR+ cells presented here. As of data cutoff at 23/10/2022, the median follow-up (mFU) was 146 days (range, 18-314), with a median progression-free survival (mPFS) and overall survival (mOS) not reached yet at the higher dose.

The over all response rate for all cohorts was 83%, with a 90% response rate in the 800x10^6 cohort. Both OS% and PFS% were significantly better in the 800x10^6 cohort compared to the previous cohorts.

MM Patients

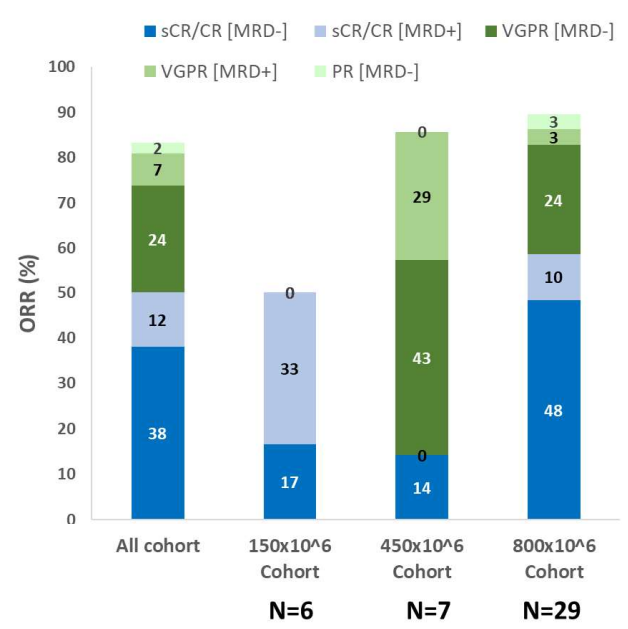
sCR/CR [MRD-]
sCR/CR [MRD+]
VGPR [MRD-]
VGPR [MRD+]
PR [MRD-]
ORR (%)
All cohort
150x10^6 Cohort
450x10^6 Cohort
800x10^6 Cohort
N=6
N=7
N=29

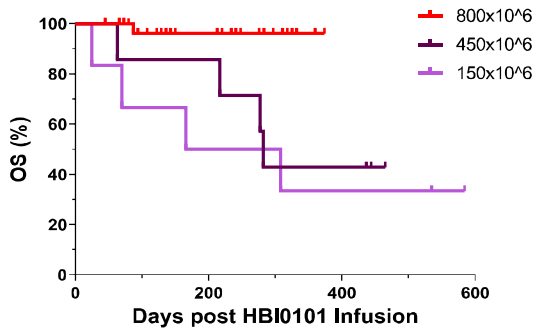
800x10^6
450x10^6
150x10^6
OS (%)
Days post HBI0101 Infusion

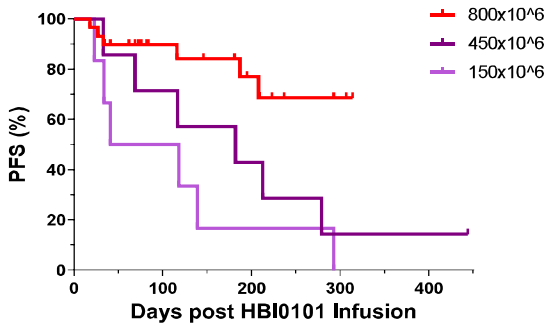
800x10^6
450x10^6
150x10^6
PFS (%)
Days post HBI0101 Infusion

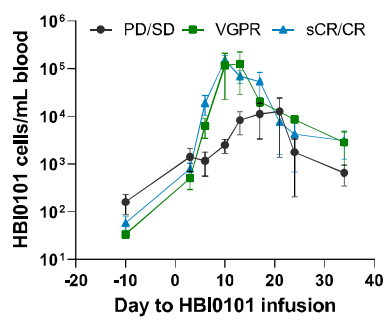
PD/SD
VGPR
sCR/CR
HBI0101 cells/mL blood
Day to HBI0101 infusion

HBI0101 *in-vivo* kinetics in HBI0101-treated MM patients, classified by stable disease/ progression disease (SD/PD) shows correlation and significant difference in pick levels of HBI0101.

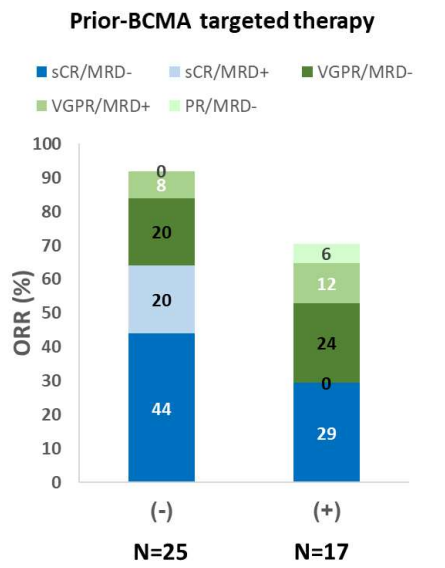
Prior-BCMA targeted therapy
sCR/MRD-
sCR/MRD+
VGPR/MRD-
VGPR/MRD+
PR/MRD-
ORR (%)
(-)
(+)
N=25
N=17

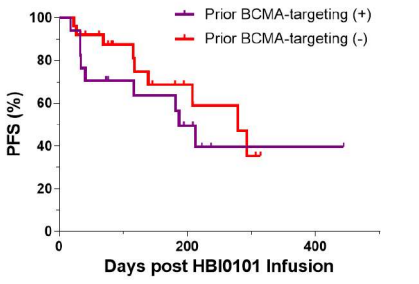
Prior BCMA-targeting (+)
Prior BCMA-targeting (-)
PFS (%)
Days post HBI0101 Infusion

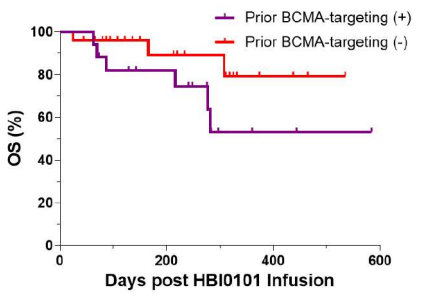
Prior BCMA-targeting (+)
Prior BCMA-targeting (-)
OS (%)
Days post HBI0101 Infusion

Previous exposure to anti BCMA therapy resulted in less favorable response rates (71% vs. 92%).

AL Patients

Patients Characterization

	Patient 1	Patient 2	Patient 3	Patient 4	Patient 5
Age	64	58	82	63	64
Gender	Male	Female	Male	Male	Male
Organ involvement	Cardiac, Renal, Autonomic	Cardiac, Renal, Hepatic	Renal, GI	Cardiac, Hepatic, Lung, Soft tissue, Autonomic	Cardiac, Soft tissue, PNS
belantamab refractory	No	Yes	No	Yes	No

Efficacy data

	Patient 1	Patient 2	Patient 3	Patient 4	Patient 5
HBI0101 Dose ($x10^6$)	150	450	800	450	800
Best Response	CR	CR	CR	CR	CR
MRD negativity (10^5)	Yes	Yes	Yes	Yes	Yes
Organ response	Yes	Yes	Yes	Yes	Yes

Similar to the MM patients, treatment of AL patients suggests better and deeper response in the 800x10^6 dose. As the study progresses we continue to enroll additional AL patients.

<u>Toxicities:</u> **A**) Hematological complications for MM and AL patients. **No ICANS** incidents were recorded and while CRS occurred in 90% (38/42) of MM patients and 80% (4/5) of AL patients they were manageable. **B**) Vast majority of the CRS events started at the day of infusion. The median duration of the CRS was 2 days (range 1-7) for MM and 4 days (range 1-5) for AL patients. **C**) Tocilisumab doses were required in ≥ Grade 2 CRS events. 2 MM patients and 1 AL patient required steroids while 1 MM and 3 AL required Vasopressors.

A

	Thrombocytopenia	Anemia	Neutropenia & Lymphopenia	Febrile Neutropenia	>28 d Hypoglobulinemia
MM COHORT					
Grade 1-2	7/42	15/42			30/42
Grade 3-4	23/42	25/42	42/42	27/42	
AL COHORT					
Grade 1-2	2/5	1/5			3/5
Grade 3-4	1/5	1/5	5/5	4/5	

B

	MM Cohort			AL Cohort	
Day Onset / CRS Grade	Grade 1 (14/42)	Grade 2 (23/42)	Grade 3 (1/42)	Grade 2 (1/5)	Grade 3 (3/5)
Day 0	9	20	1	1	3
Day 1	2	1			
Day 3	1	1			
Day ≥4	2	1			

C

	MM Cohort (25/42)			AL Cohort (4/5)	
Doses Toci / CRS Grade	Grade 1	Grade 2	Grade 3	Grade 2	Grade 3
None	13	-	-	-	-
1	1	13	-	1	-
2	-	7	-	1	-
3	-	1	-	1	-
4	-	2	1	-	1

Additional events: 9/42 MM patients had renal failure, 10/42 ELFT and 4/42 Bacteremia during hospitalization. No such events were recorded for the AL patients

CONCLUSION

HBI0101 CART product exhibits comparable safety and efficacy properties as the commercial anti-BCMA CART products reported in the literature (ABECMA™), with a clinically manageable safety profile and promising *in-vivo* efficacy. Hadassah Medical Center's experience testifies of the feasibility of a decentralized approach for point-of-care CART manufacture to supply the local demand.

REFERENCES

1. *Harush O, Asherie N, Kfir-Erenfeld S, Adler G, Barliya T, Assayag M, Gatt ME, Stepensky P, Cohen CJ. Preclinical evaluation and structural optimization of anti-BCMA CAR to target multiple myeloma. Haematologica. 2022 Oct 1;107(10):2395-2407. doi: 10.3324/haematol.2021.280169. PMID: 35354252;*
2. *Kfir-Erenfeld S, Asherie N, Grisariu S, Avni B, Zimran E, Assayag M, Sharon TD, Pick M, Lebel E, Shaulov A, Cohen YC, Avivi I, Cohen CJ, Stepensky P, Gatt ME. Feasibility of a Novel Academic BCMA-CART (HBI0101) for the Treatment of Relapsed and Refractory AL Amyloidosis. Clin Cancer Res. 2022 Dec 1;28(23):5156-5166. doi: 10.1158/1078-0432.CCR-22-0637. PMID: 36107221.*
3. *Asherie N, Kfir-Erenfeld S, Avni B, Assayag M, Dubnikov T, Zalcman N, Lebel E, Zimran E, Shaulov A, Pick M, Cohen Y, Avivi I, Cohen C, Gatt ME, Grisariu S, Stepensky P. Development and manufacturing of novel locally produced anti-BCMA CART cells for the treatment of relapsed/refractory multiple myeloma: phase I clinical results. Haematologica. 2022 Oct 6. doi: 10.3324/haematol.2022.281628. Epub ahead of print. PMID: 36200421.*

5th European CAR T-cell Meeting